Case Study of Long-Term Safety, Tolerability, And Hemodynamic Response of PB1046, A Sustained-Release Analogue for Vasoactive Intestinal Peptide (VIP), in an Adult Subject With Pulmonary Arterial Hypertension (PAH)

Presented at the
14th PVRI World Congress,
Lima, Peru
January 2020

Raymond L. Benza[1], Sumita Paul[2], Murali Chakinala[3], Priscilla Correa[1], John Lee[2], and James White[4]

[1] Allegheny Health Network, [2] PhaseBio Pharmaceuticals, [3] Washington University School of Medicine, [4] University of Rochester Medical Center

Background:

- **PB1046 is a first-in-class vasoactive intestinal peptide (VIP) analogue under development for the treatment of PAH (Figure 1a):**
 - VIP's biologic effects are mediated by two G-protein-coupled receptors - VPAC1 and VPAC2 receptors.
 - VPAC2 in the pulmonary vasculature may play a role in PAH pathogenesis.
 - PB1046 is 100x more selective for VPAC2 compared to VPAC1 (Figure 1b).
- **Molecular Structure of PB1046:**
 - PB1046 is a 634 amino acid recombinant fusion protein expressed in *Escherichia coli.*
 - Human VIP is at the N-terminus with a physiologically inert polymeric elastin-like polypeptide (ELP) sequence at the C-terminus, resulting in a >60 hr half-life of the VIP analogue (Figure 1).
 - The VIP moiety comprises the first 28 amino acids of mature human VIP with a single additional methionine at the N-terminus to confer VPAC2 selectivity.
- **CardioMEMS™ HF System:**

 The safety and accuracy of the CardioMEMS™ HF System, which monitors pulmonary artery (PA) pressure from a sensor implanted into the PA, has been previously documented, along with correlations with Swan-Ganz measurements and echocardiography.

Methods:

- The multi-dose safety, PK, and VIP-based pharmacodynamic effects of PB1046 were evaluated in an open-label, multi-dose Phase 1 pilot study in PAH patients who have a permanently implanted CardioMEMS™.
- PB1046 was administered subcutaneously once-weekly for 8 weeks (extended due to subjective improvements) at dose levels previously tested and shown to be safe.

Results:

- Three patients presented previously (PVRI 2018) completed the study with no related serious adverse events, symptomatic hypotension, or syncope. PB1046 was well tolerated with only mild injection site erythema.
- In 2 patients, hemodynamic monitoring showed reduced mean PA pressure and total pulmonary resistance with increased stroke volume and cardiac output and stable heart rate. One patient elected long-term maintenance therapy and remained on PB1046 for >18 months total.
- Long-term PB1046 therapy demonstrated clinically meaningful improvement in all hemodynamic parameters assessed, which remained stable throughout the treatment period (Figure 2).
- After completing approximately 1 year of therapy, the patient's dose was tapered gradually prior to discontinuation. After each incremental dose reduction, hemodynamic perturbation was observed followed by stabilization (Figure 3, red boxes).
- Withdrawal of therapy led to marked reversion of hemodynamic parameters to pretreatment levels approximately 3 months post-final dose of PB1046 (Figure 3a and b).

Mechanism of PB1046– Slow Release and Prolonged Half-Life:

Figure 1a. PB1046: Long-lasting VIP analogue

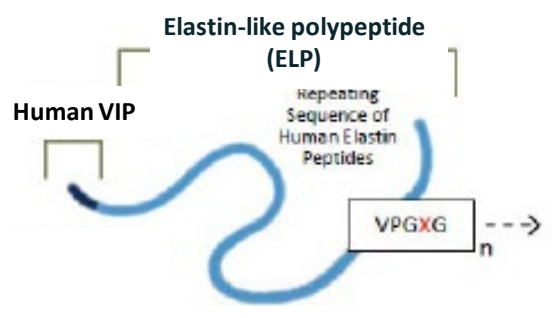

Elastin-like polypeptide (ELP)
Human VIP
Repeating Sequence of Human Elastin Peptides
VPGXG
n

Effective half life > 60 hours

Figure 1b. PB1046: VPAC2 selective

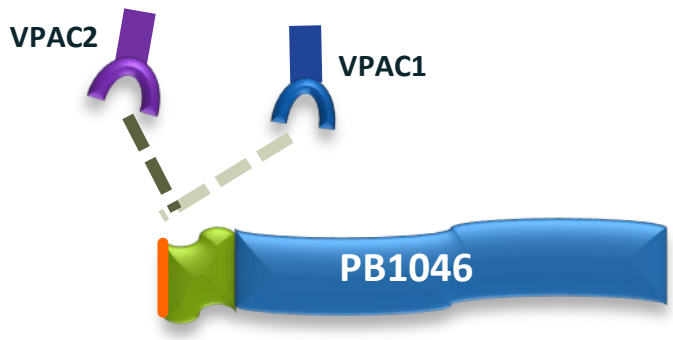

VPAC2
VPAC1
PB1046

100X VPAC2 selectivity

Figure 2. PAH patient initial hemodynamic responses

PB1046 effect on mean PA pressure

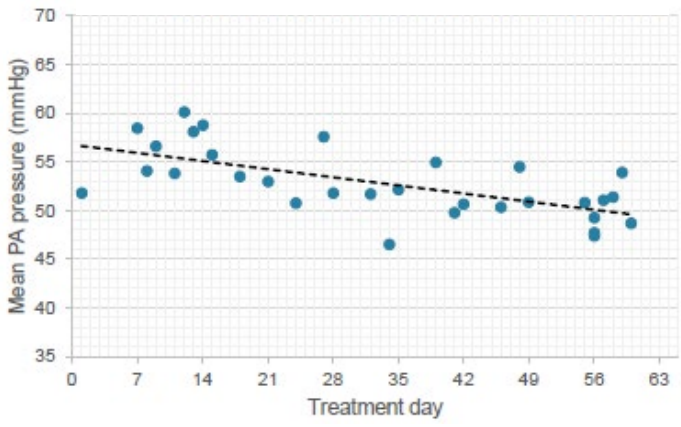

Mean PA pressure (mmHg)
70
65
60
55
50
45
40
35
0
7
14
21
28
35
42
49
56
63
Treatment day

PB1046 effect on total pulmonary resistance

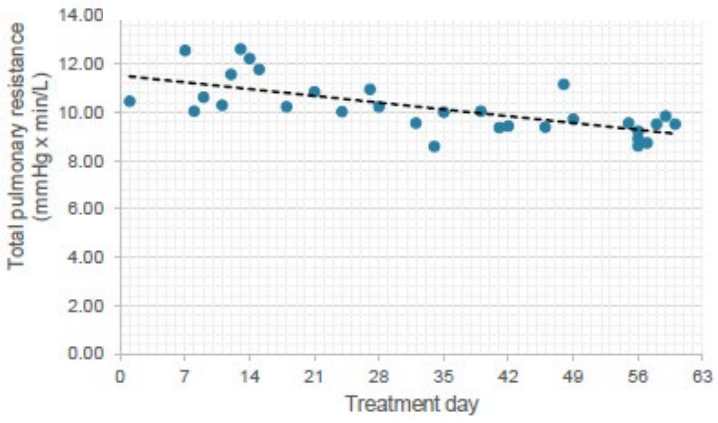

Total pulmonary resistance (mmHg x min/L)
14.00
12.00
10.00
8.00
6.00
4.00
2.00
0.00
0
7
14
21
28
35
42
49
56
63
Treatment day

Figure 3a. Long-term hemodynamic response of PB1046

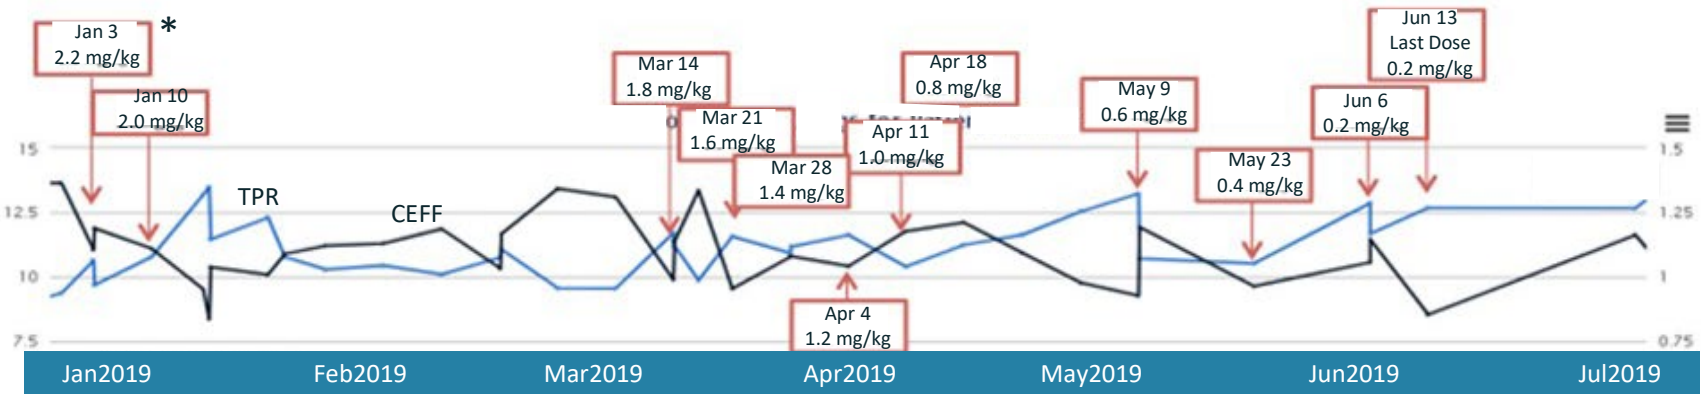

Jan 3 2.2 mg/kg
*
Jan 10 2.0 mg/kg
Mar 14 1.8 mg/kg
Mar 21 1.6 mg/kg
Mar 28 1.4 mg/kg
Apr 4 1.2 mg/kg
Apr 11 1.0 mg/kg
Apr 18 0.8 mg/kg
May 9 0.6 mg/kg
May 23 0.4 mg/kg
Jun 6 0.2 mg/kg
Jun 13 Last Dose 0.2 mg/kg
TPR
CEFF
15
12.5
10
7.5
1.5
1.25
1
0.75
Jan2019
Feb2019
Mar2019
Apr2019
May2019
Jun2019
Jul2019

*Red boxes indicate timing of dose reduction

TPR= Total pulmonary resistance
CEFF=Efficiency

Figure 3b. Reversal of PB1046 effect after study drug discontinuation

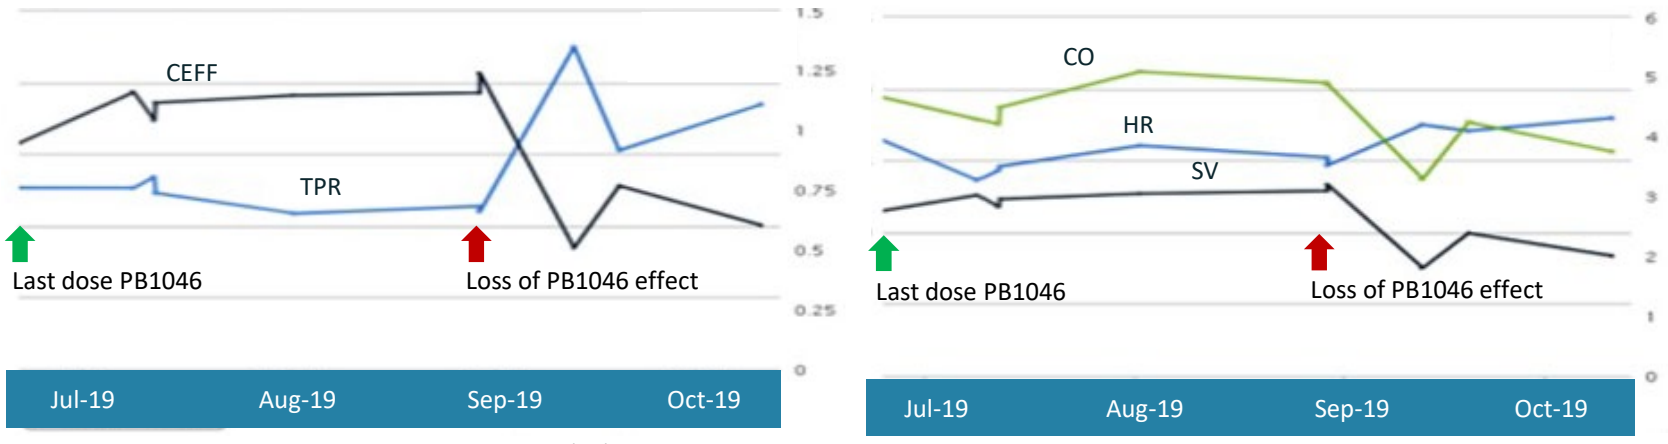

CEFF
TPR
Last dose PB1046
Loss of PB1046 effect
1.5
1.25
1
0.75
0.5
0.25
0
Jul-19
Aug-19
Sep-19
Oct-19
CO
HR
SV
Last dose PB1046
Loss of PB1046 effect
6
5
4
3
2
1
0
Jul-19
Aug-19
Sep-19
Oct-19

TPR= Total pulmonary resistance
CEFF=Efficiency

CO = Cardiac output
HR = Heart rate
SV = Stroke volume

Conclusions:

- In a case study of a PAH patient treated for >18 months with PB1046, long-term, sustained improvements in pulmonary hemodynamics that gradually reversed post-discontinuation of study drug suggest possible disease modifying effects of PB1046 and support ongoing investigation of PB1046 as a novel VIP-based therapy for PAH patients.
- A randomized, double-blind, parallel-group Phase 2b PAH study is ongoing to assess 16 weeks of PB1046 treatment added to standard of care therapy for NYHA/WHO functional class II and III PAH patients.

Acknowledgements:
We thank the patients who participated in this study and the research teams and investigators who made this study possible.